Screening For ATP Citrate Lyase Inhibitors Using Mass Spectrometry

Eliza Peterson[1], Kurt Morgenstern[1], Barbara Grubinska[2], Can Ozbal[3], William LaMarr[3]
[1]Lead Discovery Amgen, Inc. Cambridge, MA [2]Oncology Amgen, Inc. Cambridge, MA and [3]Biotrove, Inc., Woburn, MA

Introduction

ATP-citrate lyase (ACL), a cytosolic ~400 kDa homotetrameric enzyme, catalyzes the synthesis of acetyl-CoA which is a requisite building block for *de novo* lipogenesis. Acetyl-CoA is an intermediate for malonyl-CoA and mevaolonate, the committed substrates for lipogenesis. Increased levels of acetyl-CoA and the resulting *de novo* lipogenesis occurs downstream of multiple oncogenic pathways and has been shown to correlate with tumor aggressiveness [1,2]. Previous studies have demonstrated that ACL inhibition by RNAi or small molecule inhibitors dramatically reduces *in vitro* tumor cellular proliferation and tumor growth *in vivo* [3]. These results implicate ACL as a viable point of therapeutic intervention for many human cancers.

In this study, we report the results of a high throughput RapidFire™ mass spectrometry assay to identify, characterize and validate ACL inhibitors. We further present our phosphopeptide mapping of protein kinase A (PKA) treated ACL using FT-Orbitrap® mass spectrometry. Moreover, the high resolution of FT-Orbitrap® was utilized for a novel proximity assay that measures the direct detection of ACL activity in cells. Using several different applications of mass spectrometry for this ACL program, we emphasize the value of mass spectrometry technology for protein and target biology programs.

[1] Ookhtens, et al. 1984, *Am J Physiol*. [2] Savine, et al. 1967, *Cancer Res*. [3] Thompson, et al. 2005, *Cancer Cell*.

ACL Cellular Function

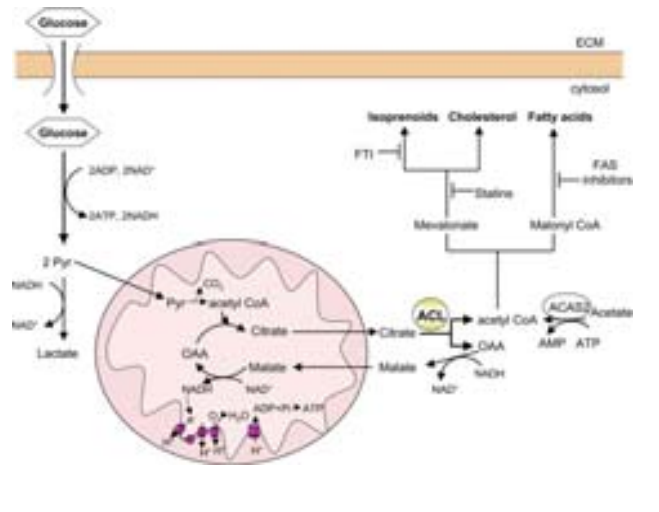
Glucose
ECM
cytosol
Isoprenoids Cholesterol Fatty acids
FTI
FAS inhibitors
Statins
Mevalonate
Malonyl CoA
2 Pyr
Lactate
Pyr
acetyl CoA
Citrate
OAA
Malate
ACL
ACAS2
Acetate
AMP ATP

ACL Enzymatic Reaction

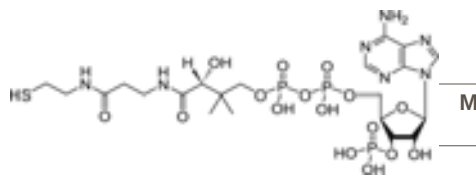

Substrate

Mass =767 amu CoA

ACL + ATP/Coenzyme A/Citrate

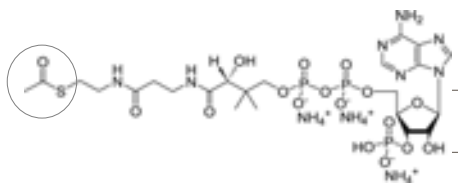

Product

Mass =810 amu Acetyl CoA

RapidFire™ Technology

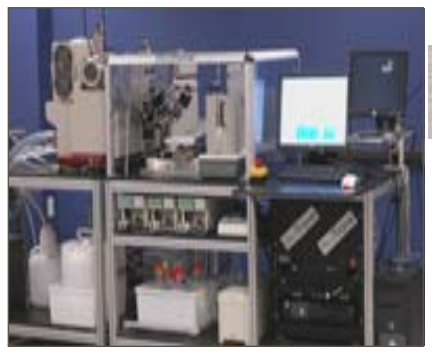

RapidFire™ Highlights

- **Direct detection of substrate/product**
- **Substrate can be lipid, peptide, metabolite, oligonucleotide, etc.**
- **fmol sensitivity**
- **6 sec/well**
- **No fluorescence interference**

RapidFire™ ACL Assay Summary

1 nM (final) ACL Enzyme in Assay Buffer
(50 mM Tris, 15mM NaCl, 10% Glycerol, 1mM MgCl2 0.05% BSA, 1mM DTT)

35 µL of enzyme + 1 µL of 1mM compound in DMSO

Pre Incubate 20 min
Room Temperature

Initiate rxn with 4 µL of 10X CoA/ATP/Citrate final concentration of 2/3 K_m
Coenzyme A K_m = 6 µM
ATP K_m = 15 µM
Citrate K_m = 75 µM

Incubate 60 min
Room Temperature

Quench with 4.5 µL 10% formic acid

Inject 5 µL, C18 cartridge
RapidFire™ (6 sec/well)

API 4000™ LC/MS/MS System
Triple Quadrupole Mass Spectrometer
Negative ionization and MRM Analysis

Q1: Full scan CoA - 766 amu (substrate)
Acetyl CoA - 810 amu (product)

Q2: Fragments precursor ions

Q3: Product ions at 408 amu

% Substrate Conversion

RapidFire™ Raw Data View

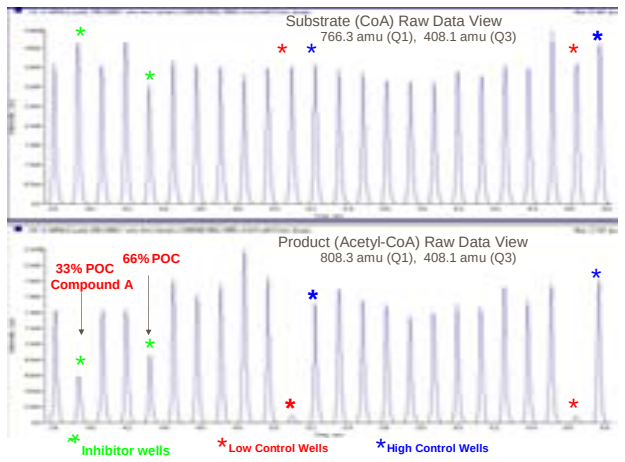
Substrate (CoA) Raw Data View
766.3 amu (Q1), 408.1 amu (Q3)
Product (Acetyl-CoA) Raw Data View
808.3 amu (Q1), 408.1 amu (Q3)
33% POC
Compound A
66% POC
Inhibitor wells
Low Control Wells
High Control Wells

Data Analysis: % Conversion= AcCoA (AUC) / AcCoA (AUC) + CoA (AUC) x 100

RapidFire™ Screening Results

An optimal RapidFire™ enzyme assay gave Z-factors between 0.70-0.96 and was used to screen 43,600 compounds in single point and more than 300 compounds in 10-point dose response follow-up titrations.

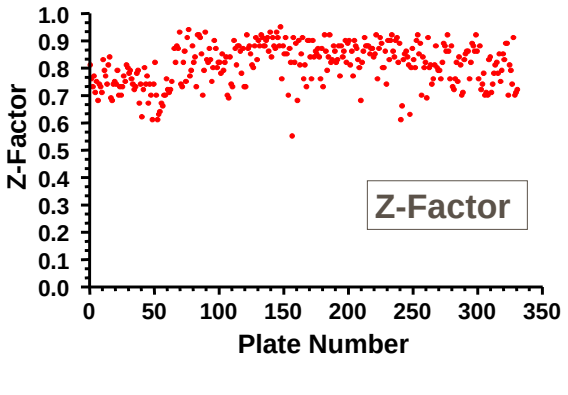
Z-Factor
Plate Number
Z-Factor

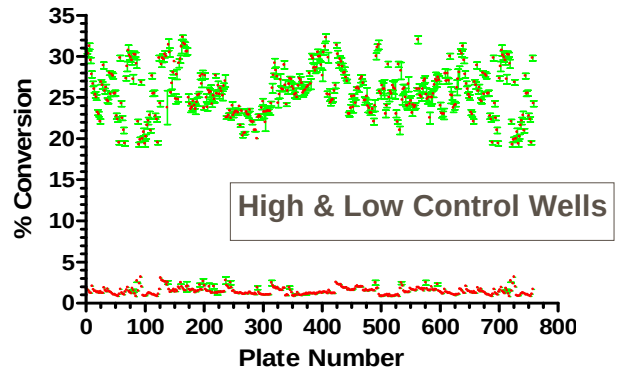
% Conversion
Plate Number
High & Low Control Wells

Leads from RapidFire™ Screen

Compound A

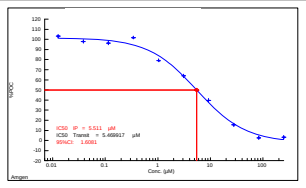

RapidFireTM HT-MS (IC50, µM)	5.9 ± 1.3, n=3 (DMSO, QC=95%)
Transcreener (IC50, µM)	6.4 ± 2.1, n=2 (Solid, QC=100%)

Compound B

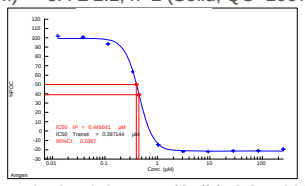

RapidFire™ HT-MS (IC50, µM)	0.43 ± 0.2, n=11 (Solid, QC=100%)
Transcreener (IC50, µM)	0.38 ± 0.04, n=2 (DMSO, QC=95%)

RapidFire™ Reversibility Studies

RapidFire™ Reversibility Study Protocol

3 µM ACL(-citrate lot) +/- 100 µM Inhibitor → Pre Inc. 20 min/RT → Dilute 1:3,000 → 33 nM [Inh] Final (10X below IC50) 1 nM ACL Final → a. Wells + [I] b. Wells – [I] → Add 2/3 K_m Ligands to Initiate Rxn → Time Course

Compound A Reversibility
No Pre Inc. in Wells

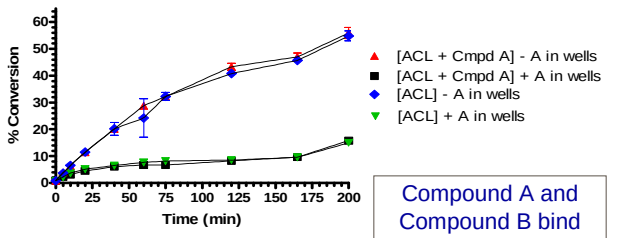
% Conversion
Time (min)
[ACL + Cmpd A] - A in wells
[ACL + Cmpd A] + A in wells
[ACL] - A in wells
[ACL] + A in wells

Compound A and Compound B bind reversibly to ACL

Compound B Reversibility
With Pre Inc. in Wells

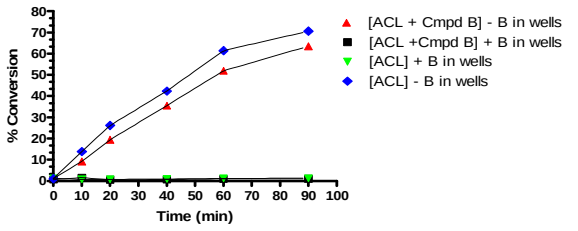
% Conversion
Time (min)
[ACL + Cmpd B] - B in wells
[ACL +Cmpd B] + B in wells
[ACL] + B in wells
[ACL] - B in wells

FT-Orbitrap® Technology

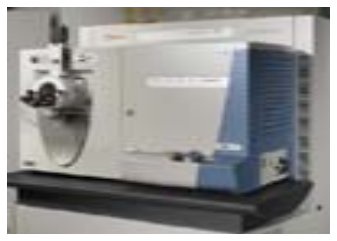

Capable of 2-5 ppm Mass Accuracy at 125,000 Resolution

Characterization of +/- PKA ACL

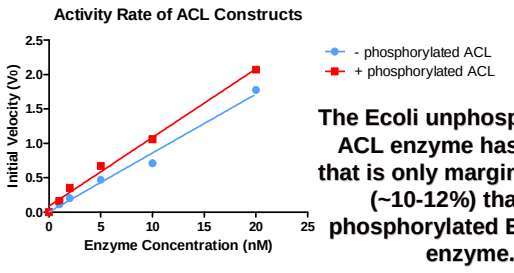
Activity Rate of ACL Constructs
Initial Velocity (Vo)
Enzyme Concentration (nM)
- phosphorylated ACL
+ phosphorylated ACL

The Ecoli unphosphorylated ACL enzyme has activity that is only marginally lower (~10-12%) than the phosphorylated Ecoli ACL enzyme.

In contrast, Potapova, et al. reported a 6-fold increase in Ecoli expressed ACL phosphorylated by PKA [Potapova, et al. 2000, *Biochemistry*].

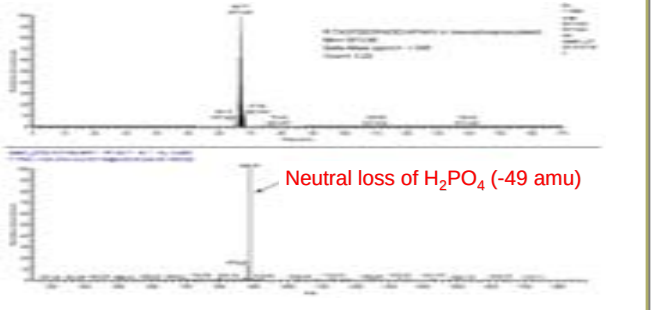
Neutral loss of H2PO4 (-49 amu)

Phosphomapping of +/- PKA Ecoli ACL enzymes using FT-Orbitrap® MS found a tryptic peptide corresponding to residues 453-569 that is monophosphorylated (based on neutral loss of H_2PO_4)

FT-Orbitrap® Cell Assay Protocol Summary

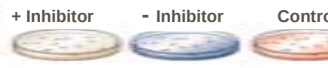
+ Inhibitor
- Inhibitor
Control

~1 X 10^7 A549 Cells

Cell Lysates

C18 Microcolumn Clean-up

Capillary Hydrophilic C18 Column

FT-Orbitrap®

FT-Orbitrap® Raw Data View and Cell Assay Data

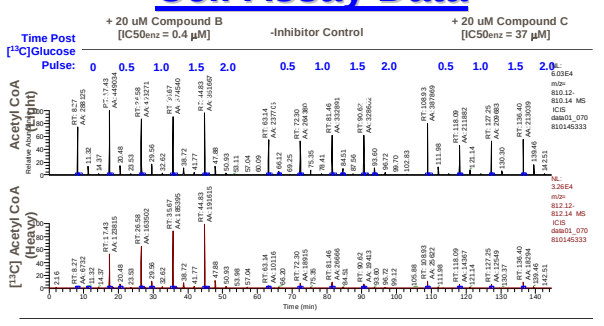
+ 20 uM Compound B [IC50enz = 0.4 µM]
-Inhibitor Control
+ 20 uM Compound C [IC50enz = 37 µM]
Time Post [13C]Glucose Pulse:
Acetyl CoA (Light)
[13C] Acetyl CoA (Heavy)
Time (min)

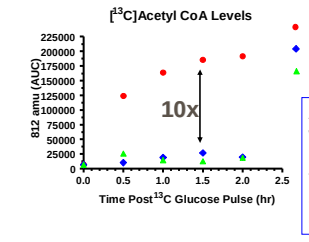
[13C]Acetyl CoA Levels
812 amu (AUC)
Time Post 13C Glucose Pulse (hr)
10x
[13C]Acetyl CoA (+) Compound B
[13C]Acetyl CoA (-) Inhibitor
[13C]Acetyl CoA (+) Compound C

A 10-fold increase in [13C] Acetyl CoA was observed in cells treated for 8 hours with 20 µM Compound B. A ~3-fold change was observed in the DMSO and Compound C negative inhibitor control cells when compared to 0 hrs.

Hypothesis of ACL Cellular Function Upon Small Molecule Inhibition

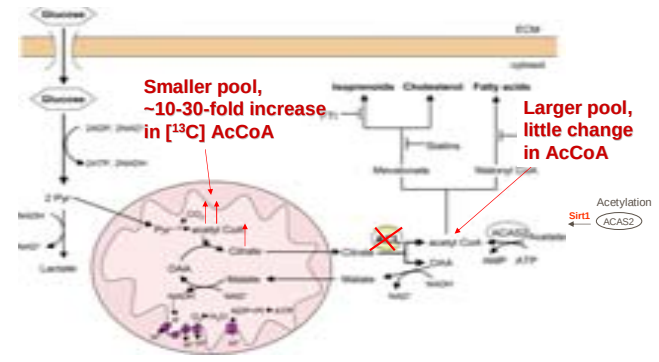
13C Glucose Pulse
Smaller pool, ~10-30-fold increase in [13C] AcCoA
Larger pool, little change in AcCoA
Acetylation
Sirt1
ACAS2

A549 Cellular Dose Response for Compound B
FT-MS Cell Assay for ACL

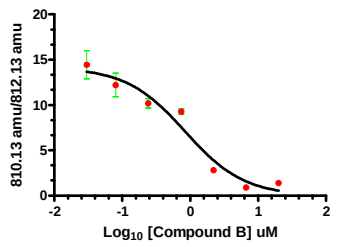
810.13 amu/812.13 amu
Log10 [Compound B] uM

RapidFire™ HT-MS (IC50, µM)	0.43 ± 0.2, n=11 (Solid, QC=100%)
Transcreener (IC50, µM)	0.38 ± 0.04, n=2 (DMSO, QC=95%)
MS Cellular Assay (EC50, µM)	1.20 ± 0.36, n=4 (Solid, QC=100%)

Conclusion

Using RapidFire™ technology to identify potential ACL inhibitors, we have screened more than 43,600 compounds in single point and more than 300 IC50 follow-ups. It has been showed that the RapidFire™ platform is ideal for identifying and verifying lead compounds such as Compound A and Compound B. RapidFire™ technology was also used to look at SAR around these most potent compounds as well as investigate reversibility and probe the mechanisms of inhibition.

Using both RapidFire™ and FT- Orbitrap® mass spectrometry to characterize PKA treated ACL we phospho-residue mapped and assayed under Vmax conditions, though report only marginally lower activity with the unphosphorylated ACL enzyme as opposed to Potapova et.al. who report a 6-fold increase in ACL phosphorylated by PKA.

In addition, the high resolution of FT-Orbitrap® enabled the direct analysis of ACL activity in cells and demonstrates compensatory increase in cellular AcetylCoA levels upon inhibition.

Acknowledgments

We would like to thank Doug Saffran, Linda Epstein, Yan Gu, Stephanie Geuns-Meyer, Yichin Liu, Leanne Zalameda, Antonio Oliveri-dos-Santos and John Newcomb for their support with this project.