THE USE OF HIGH THROUGHPUT MASS SPECTROMETRY (HTMS) IN DRUG DISCOVERY

Roger J. Schilling*, Nicole Rauccio, Andrea Quercia, Zane Wenzel, Claire Steppan, Stephan Orena, Robert Garofalo, and Thomas McLellan
Pfizer Global Research & Development, Groton Laboratories, Pfizer Inc, Groton, CT 06340

WHAT IS THE BIOTROVE HTMS?

- BioTrove's ultra High Throughput Mass Spectrometry (HTMS) system consists of a proprietary computer-controlled fluidic robot interfaced with a triple-quadrupole mass spectrometer. Robotics replace conventional HPLC (high performance liquid chromatography). The sample injection and purification system used is a modified LC-MS application.
- The BioTrove system can quantify multiple analytes simultaneously. The substrate and product of an assay can be quantified simultaneously, along with one or more internal standards as necessary.
- High quality data with coefficients of variability under 10% are routinely achieved in high-throughput screening applications. Z' values typically are > 0.6.
- The purification and mass spectrometry analysis steps described above are performed in a serial manner, eliminating the need for up-front sample preparation or purification. A wide range of buffers, salts, detergents, and compounds such as ATP or NADPH are fully compatible the BioTrove HTMS system.

WHAT IS THE BIOTROVE HTMS: INSTRUMENTATION

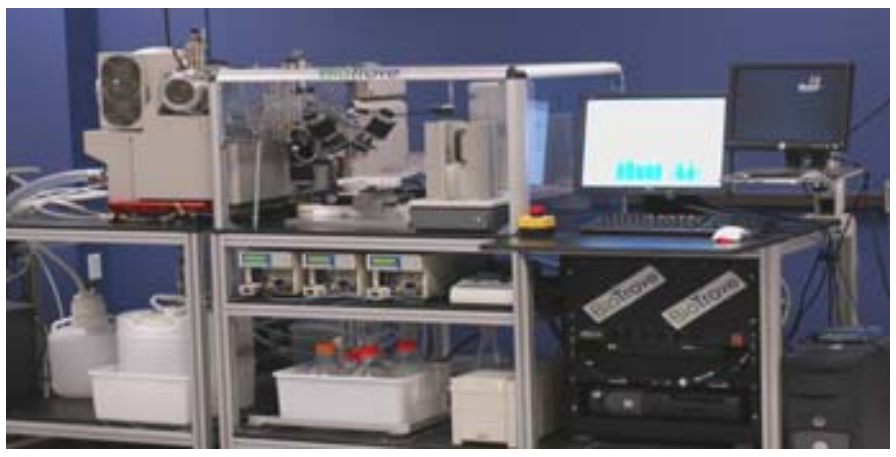

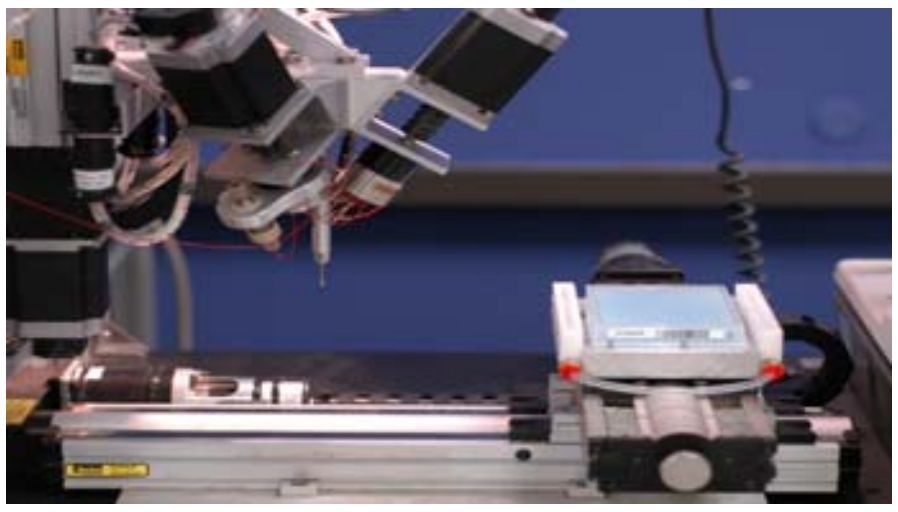

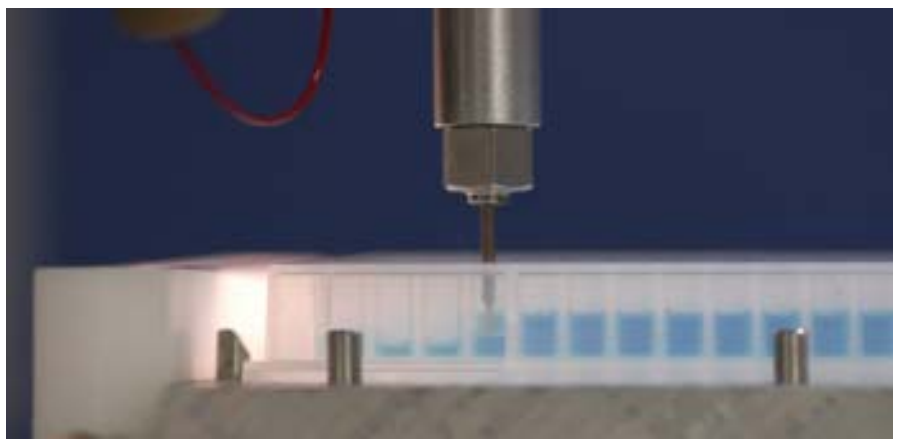

AN EXAMPLE OF SAMPLE THROUGHPUT: DGAT

- 0.75 sec = sipper positioning into each well
- <u>6.25 sec = BioTrove analysis time</u>
- 7 sec = total time per well
- 0.06 min = 3.6 sec = peak width
- 18 analysis points/peak
- 44.8 min = 384 wells
- 0.4 min = 24 sec = Wash time before each plate
- <u>0.8 min = 48 sec = Time between plates (Twister moving plates)</u>
- 46 min = total time per plate
- 8 x 384-well plates/day - 4 plates/run (acquiring with Analyst software in Tune mode which freezes if running for too long)

 → *Although this is not high-throughput for HTS, it is high throughput for Mass Spec.*

 → *his technology may impact different areas of drug discovery due to its high data quality and higher throughput.*

HOW ARE WE USING HIGH THROUGHPUT MS?

- Subset Screening for **Acyl-CoA:Diacylglycerol Acyltransferase** (DGAT)
 - Assay development
 - 210 plate compound mixtures subset screen
- Hit to Lead expansion with Stearoyl CoA Desaturase (SCD)
- Assay Development for SirT1 deacetylase

SUBSET SCREENING: ACYL-COA:DIACYLGLYCEROL ACYLTRANSFERASE (DGAT) INHIBITION

DGAT catalyzes the terminal step of TG synthesis:
transfer of acyl-CoA to diacylglycerol

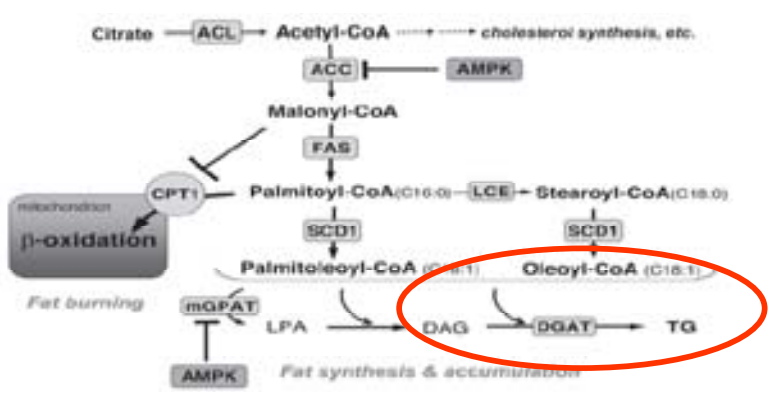
Citrate ACL Acetyl-CoA cholesterol synthesis, etc.
ACC AMPK
Malonyl-CoA
FAS
CPT1 Palmitoyl-CoA (C16:0) LCE Stearoyl-CoA (C18:0)
β-oxidation
SCD1 SCD1
Palmitoleoyl-CoA (C16:1) Oleoyl-CoA (C18:1)
Fat burning
mGPAT LPA DAG DGAT TG
AMPK
Fat synthesis & accumulation

DGAT HTMS ASSAY DEVELOPMENT

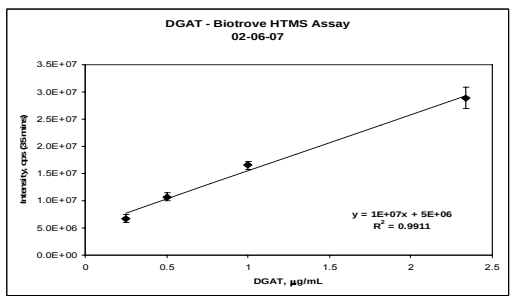
DGAT - Biotrove HTMS Assay
02-06-07
y = 1E+07x + 5E+06
R² = 0.9911
DGAT, µg/mL

There is a linear relationship between the amount of DGAT in the reaction and the amount of product formed.

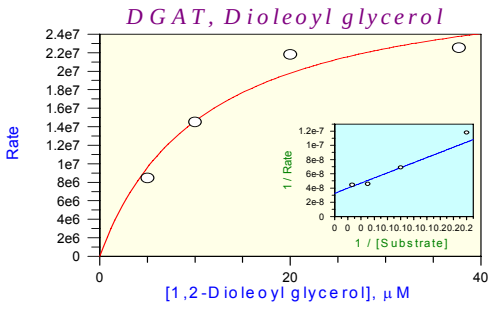
DGAT, Dioleoyl glycerol
Rate
1 / Rate
1 / [Substrate]
[1,2-Dioleoyl glycerol], µM

Parameter	Value	Std. Error
Vmax	30695092.6911	4251103.8291
Km	11.0280	4.0435

The Km of 1,2-dioleoyl glycerol is 11 µM.

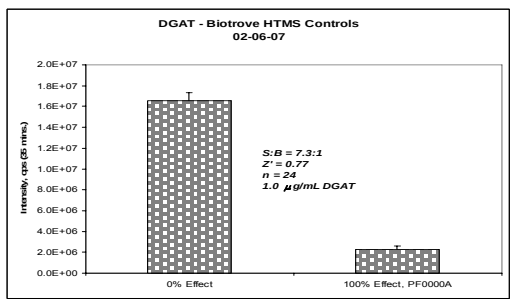
DGAT - Biotrove HTMS Controls
02-06-07
S:B = 7.3:1
Z' = 0.77
n = 24
1.0 µg/mL DGAT
0% Effect
100% Effect, PF0000A

1.0 µg/mL DGAT
35 mins, room temp.
IC_{50} = 49.1 nM
n = 24

<u>DGAT – BioTrove HTMS Assay</u>

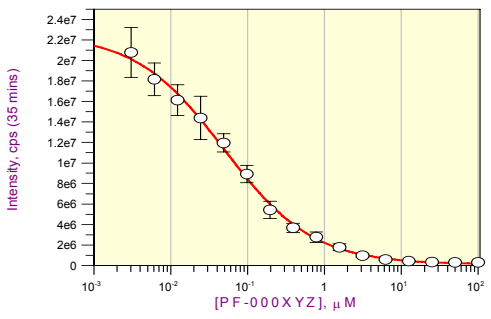
Intensity, cps (35 mins)
[PF-000XYZ], µM

Parameter	Value	Std. Error
Y Range	22485151.1849	1036376.2570
IC 50	0.0491	0.0068
Slope factor	0.7525	0.0575
Background	109402.8233	226334.0364

DGAT HTMS SCREENING WORKFLOW

513,180 compounds screened at 9 µM

↓

776 showed => 50% Inhibition (n=2), 0.15% hit rate

↓

773 were available and were tested at 9 µM

↓

96 compounds showed => 40% Inhibition (n=3), 12.4% confirmation rate

↓

50 actives demonstrated IC_{50} <= 15 µM
14 of these actives <= 1 µM
Triaged actives were confirmed in cell assays

STEAROYL COA DESATURASE

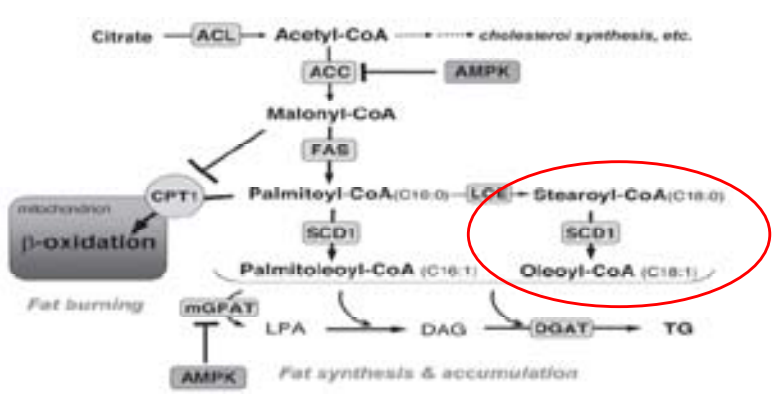
Citrate ACL Acetyl-CoA cholesterol synthesis, etc.
ACC AMPK
Malonyl-CoA
FAS
CPT1 Palmitoyl-CoA (C16:0) LCE Stearoyl-CoA (C18:0)
β-oxidation
SCD1 SCD1
Palmitoleoyl-CoA (C16:1) Oleoyl-CoA (C18:1)
Fat burning
mGPAT LPA DAG DGAT TG
AMPK
Fat synthesis & accumulation

STEAROYL-COENZYME A DESATURASE

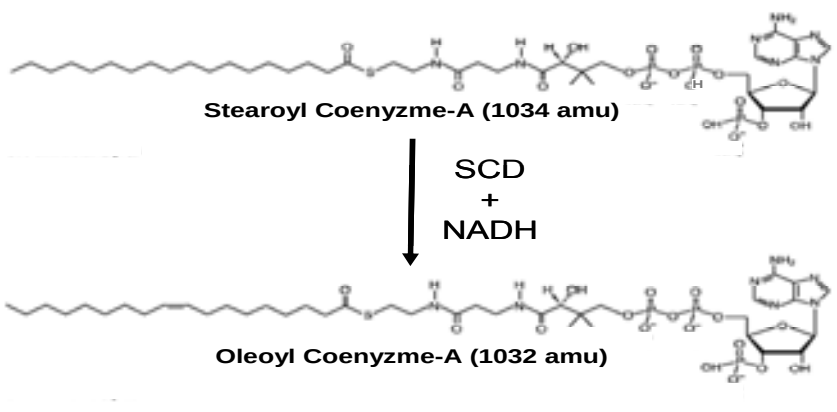
Stearoyl Coenyzme-A (1034 amu)
SCD
+
NADH
Oleoyl Coenyzme-A (1032 amu)

The BioTrove HTMS SCD assay is measuring a 2 amu difference between the desaturase substrate and product.

SCD HTMS HIT TO LEAD

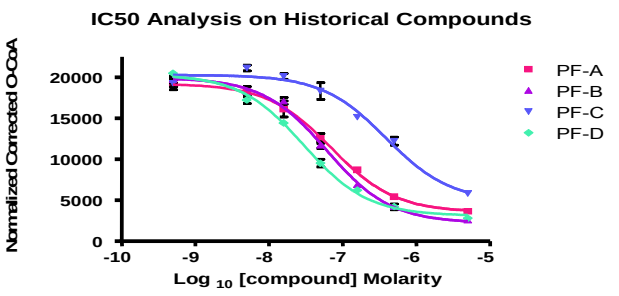
IC50 Analysis on Historical Compounds
Normalized Corrected O-CoA
Log 10 [compound] Molarity
PF-A
PF-B
PF-C
PF-D

	EC50
PF-A	7.030e-008
PF-B	6.080e-008
PF-C	3.884e-007
PF-D	2.964e-008

The HTMS SCD assay easily differentiates various IC_{50} potencies during hit to lead expansion.

SIRT1 (HOMOLOG OF YEAST SILENT INFORMATION REGULATOR 2)

- Sirtuins are 'class III' NAD⁺ dependent <u>h</u>istone <u>deac</u>etylases (HDACs)

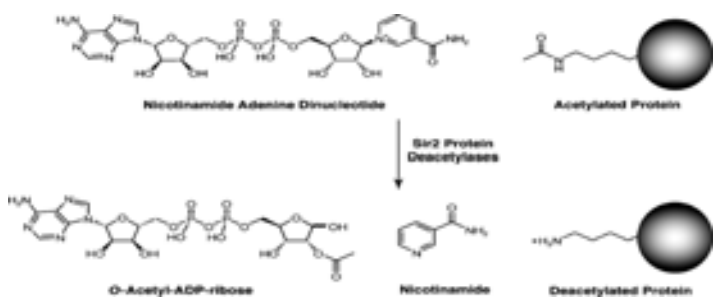
Nicotinamide Adenine Dinucleotide
Acetylated Protein
Sir2 Protein Deacetylases
O-Acetyl-ADP-ribose
Nicotinamide
Deacetylated Protein

- NAD^+-dependence links Sirt1 activity to metabolic status
- Up-regulated by caloric restriction (yeast, fly and rat); correlated to increased lifespan yeast, flies, worms and mice
- The Sirt1 activator Resveratrol extended lifespan in yeast, flies and mice
 - Shown to be Sirt1 dependent in yeast and flies

Ac-EEKGQSTSSHS-K(Ac)-Nle-STEGKEE-NH2

A.

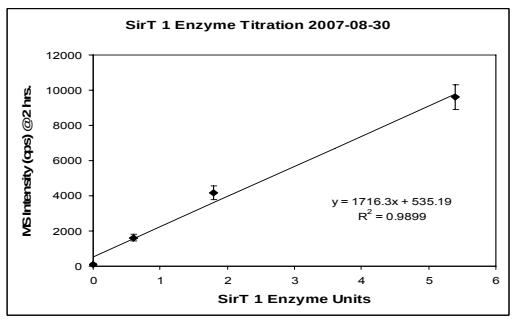
SirT 1 Enzyme Titration 2007-08-30
MS Intensity (cps) @2 hrs.
y = 1716.3x + 535.19
R² = 0.9899
SirT 1 Enzyme Units

B.

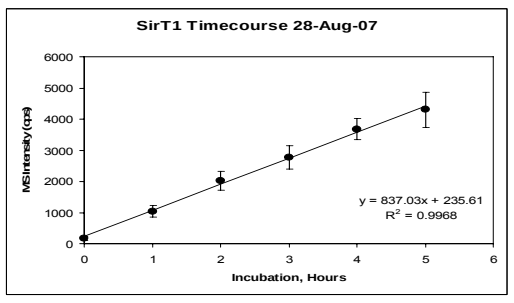
SirT1 Timecourse 28-Aug-07
MS Intensity (cps)
y = 837.03x + 235.61
R² = 0.9968
Incubation, Hours

The BioTrove HTMS SirT1 assay can detect changes in the deacetylation of a selected peptide. Figure A shows a linear relationship between the amount of product measured versus the amount of SirT1 present in the reaction.

Figure B indicates we are operating in a linear region of the reaction time.

SirT1 HTMS Km DETERMINATIONS

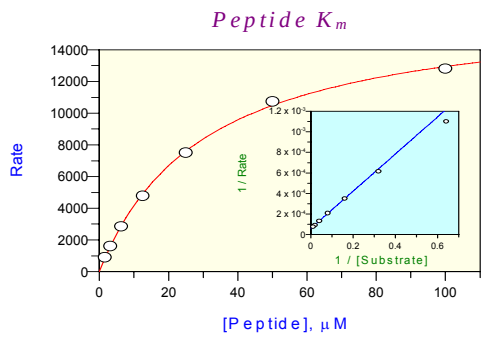
Peptide Km
Rate
1 / Rate
1 / [Substrate]
[Peptide], µM

Parameter	Value	Std. Error
Vmax	16898.6869	330.3227
Km	30.5785	1.4494

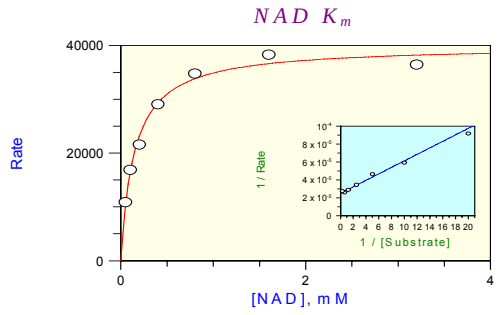
NAD Km
Rate
1 / Rate
1 / [Substrate]
[NAD], mM

Parameter	Value	Std. Error
Vmax	39939.2724	1116.5204
Km	0.1447	0.0164

SUMMARY

- The 1st generation Biotrove HTMS instrument was delivered to Pfizer Groton HTS Center of Emphasis in Sept. 2006.
- In 2007, the HTS CoE has applied the HTMS system to:
 - Assay development and Subset screening for DGAT
 - Hit to Lead exploration with Stearoyl CoA Desaturase (SCD)
 - Assay Development for SirT1
- At this time in 2008, we have expanded our impact by supporting 7 drug targets at various discovery stages.

ACKNOWLEDGEMENTS

- RJS and NR gratefully acknowledge the technical support and expert mentoring provided by Can Ozbal of BioTrove. We also thank Eliot Sugarman for supplying us with high quality liver microsomes for the DGAT studies.